A prospective, open-label, multicenter study of the efficacy, safety, tolerability, and pharmacokinetics of PlasmaCap™ IG (Immunoglobulin) in adults and children with primary immunodeficiency

Jacinthe Guindon, MSM, Mark Krause, BSc, MBA; Evolve Biologics

Introduction

Background

- Proprietary EBA technology to manufacture highly specialized plasma therapeutics was acquired in 2012. The technology allows the extraction of the native form of the protein with higher yield from each liter of plasma.

- PlasmaCap IG is the first plasma derived product manufactured using PlasmaCap EBA™. Market research was done to design the optimal formulation buffer and the product was extensively characterized by a cascade of qualified methods developed to ensure consistent product quality.

- PlasmaCap IG is a 10% liquid formulation human immunoglobulin manufactured from US source plasma for intravenous (iv) replacement therapy for primary immune deficiency diseases (PIDD) in adult and pediatric patients two years of age or older. Pre-Clinical studies were conducted in rats and rabbits to demonstrate the safety of PlasmaCap, an IND was filed in 2016 and in 2017 the Clinical trial was initiated in both the US and Canada. The purpose of this study is to investigate the efficacy, safety, tolerability and pharmacokinetic profile of PlasmaCap IG on the basis of historical control data of other 10% intravenous immunoglobulin (IGIV) products licensed in North America for the treatment of subjects with PIDD.

- The trial is expected to be complete in February 2019 (adults) and November 2020 (pediatrics). Data presented here is as of the most recent safety review in July 2018.

Pre-Clinical Data

Single Dose Intravenous Infusion Toxicity and Toxicokinetic Study with PlasmaCap IG in Sprague Dawley Rats with a 2-Week Recovery Phase

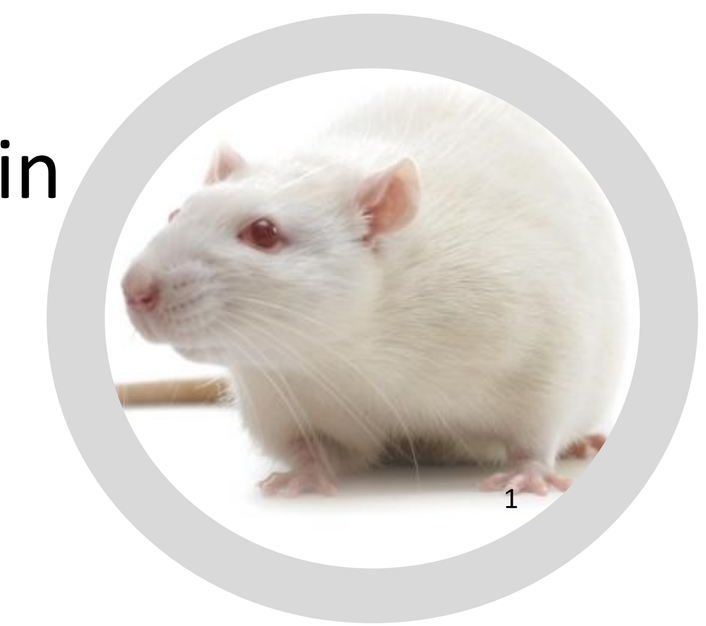

- PlasmaCap IG was well tolerated as a single dose administered via intravenous infusion at dose levels of 0, 100, 1000, or 2000 mg/kg in male and female rats.

- Based on these observations, the no observed adverse effect level (NOAEL) was considered 2000 mg/kg when given as a single intravenous infusion.

Assessment of *in vivo* Safety of PlasmaCap IG: Evaluation of Thromboembolic Adverse Events in a Rabbit Venous Stasis Model

- Statistical analysis demonstrated that the animals exposed to 900 mg/kg PlasmaCap IG in March 2017 and May 2017 exhibited thrombus scores which were indistinguishable from those animals exposed to 900 mg/kg Commercial 10% in May 2017 (4 rabbits) and January 2017 (4 rabbits: IPS Therapeutique Inc. internal GLP-validation).

- The infusion of the reference compound, Commercial 10% (900 mg/kg), caused an average thrombus score of 0.52 (n=8). The infusion of PlasmaCap IG (900 mg/kg) caused an average thrombus score of 0.73 (n=14).

Statistical analysis confirmed that PlasmaCap IG and Commercial 10% produced identical thrombus scores in the Wessler venous stasis model.

Methods

- Clinical Trial Identifier #NCT03238079

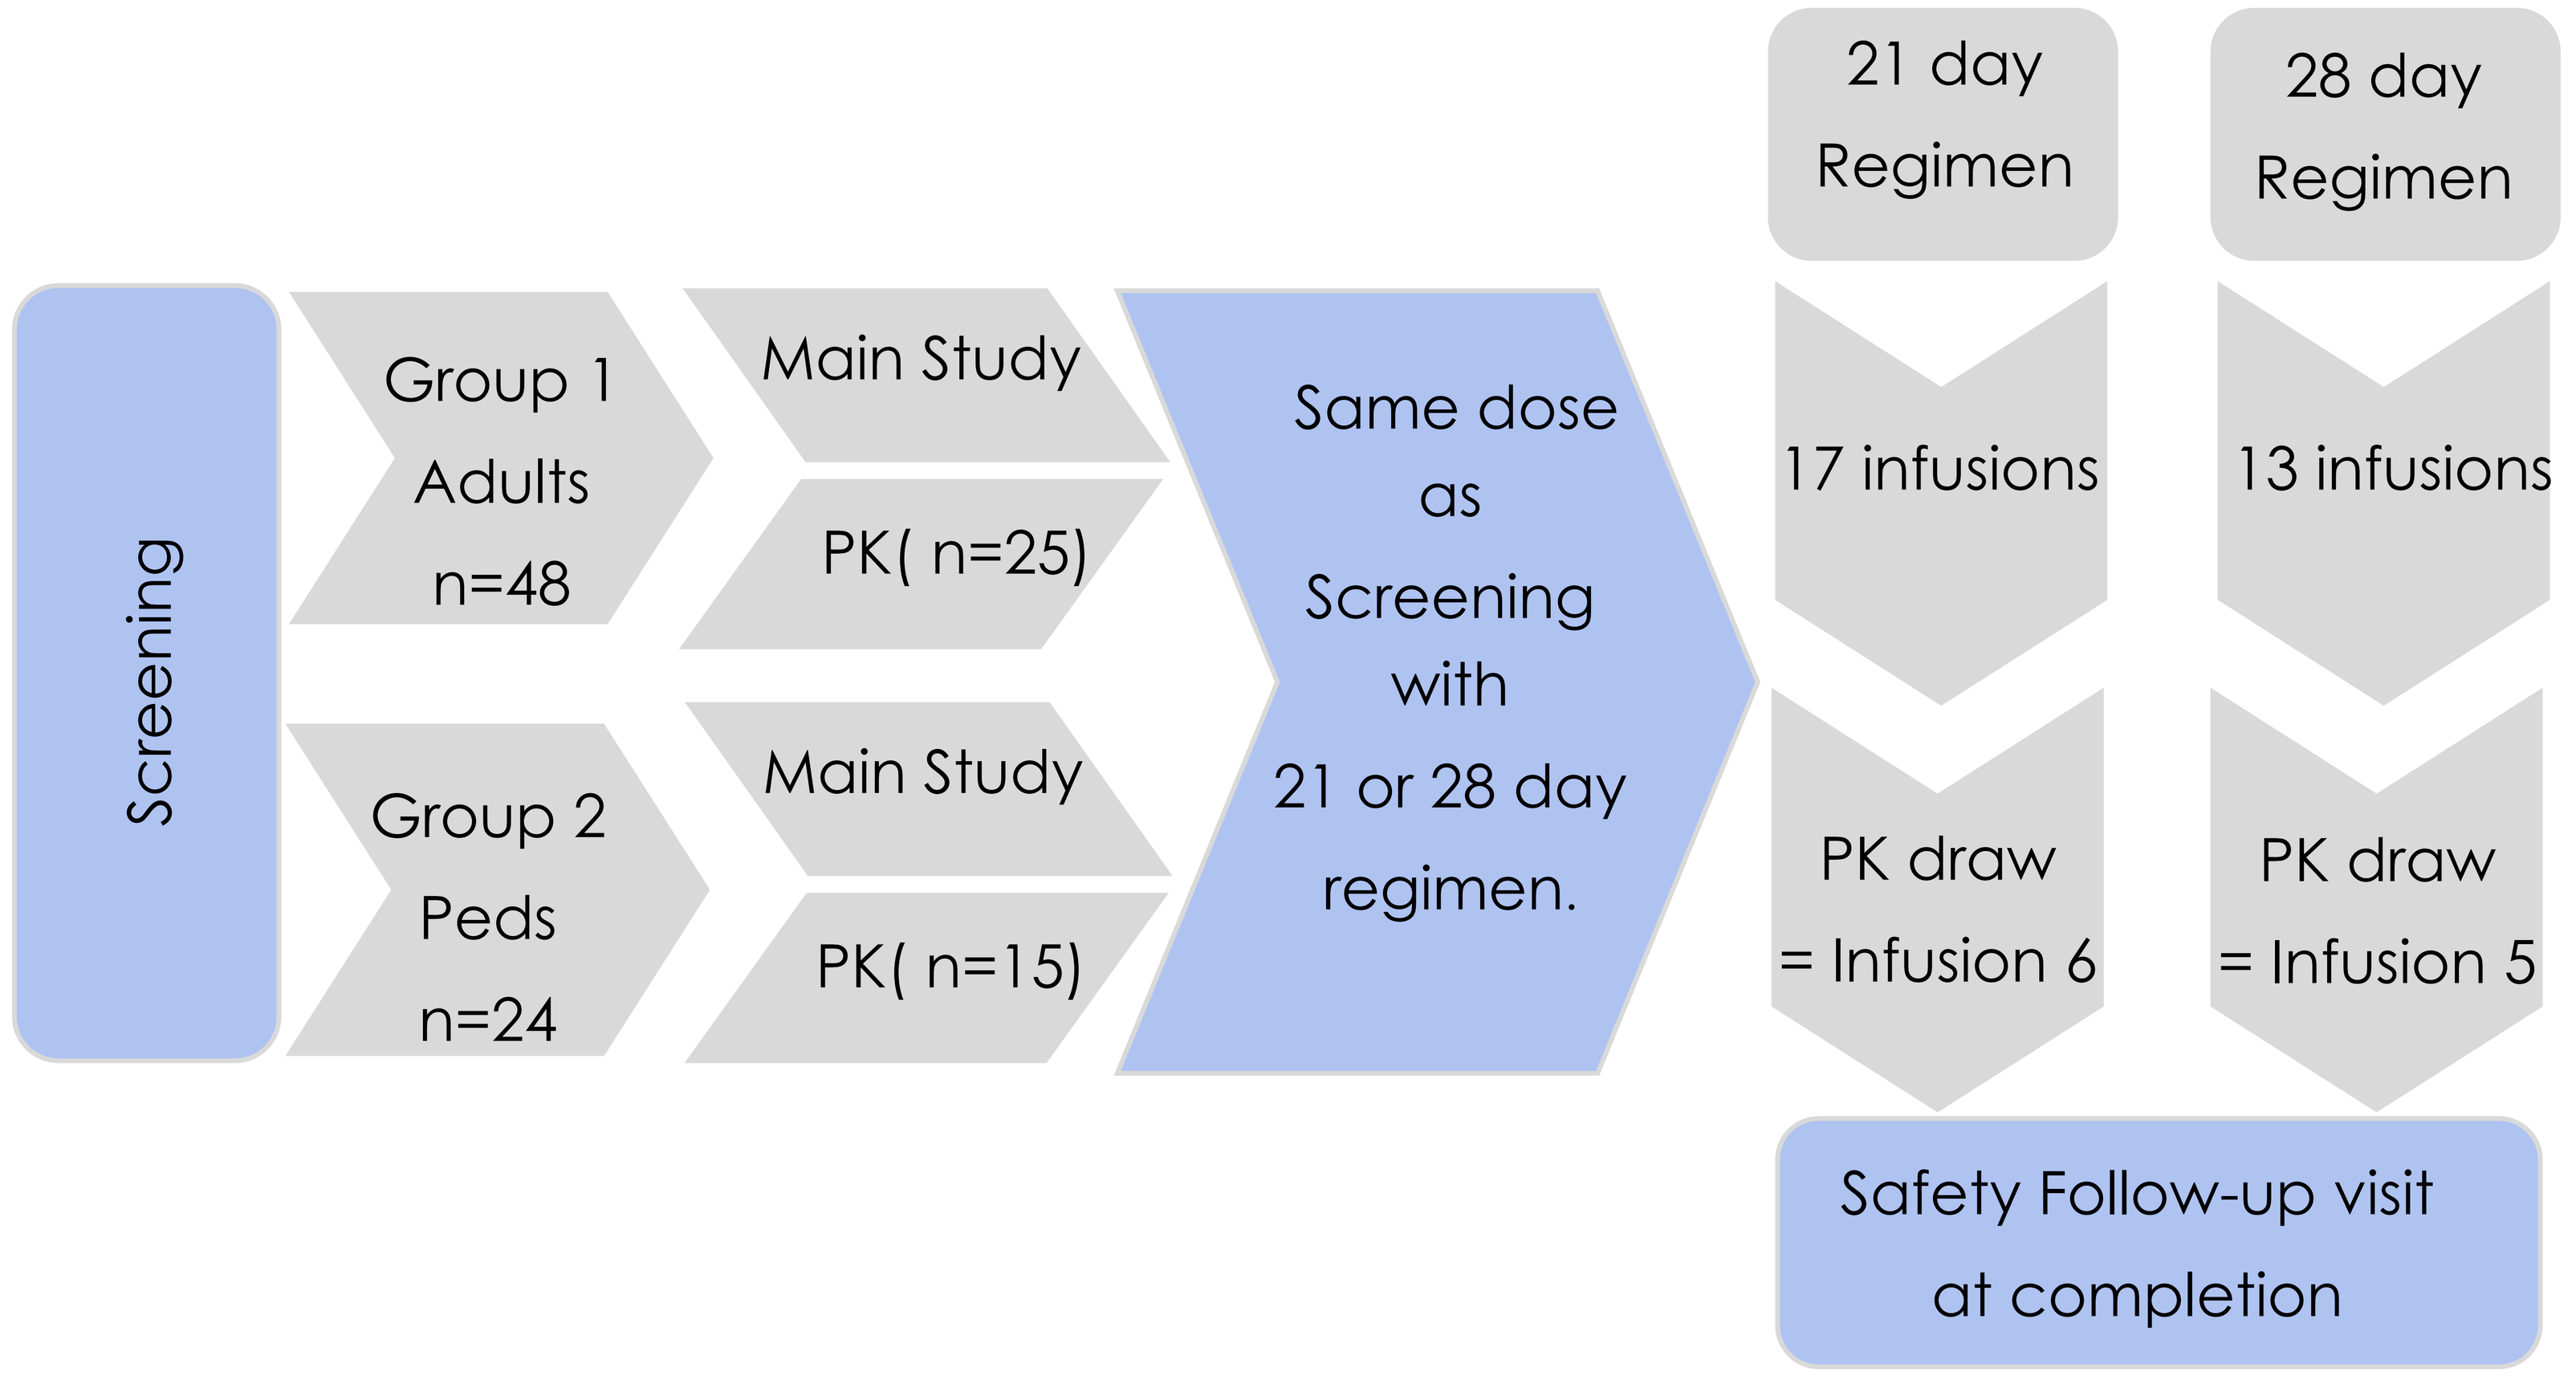
Screening
Group 1
Adults
n=48
Main Study
PK(n=25)
Group 2
Peds
n=24
Main Study
PK(n=15)
Same dose as Screening with 21 or 28 day regimen.
21 day Regimen
17 infusions
PK draw = Infusion 6
28 day Regimen
13 infusions
PK draw = Infusion 5
Safety Follow-up visit at completion

Study Outcomes Measures

Primary Outcomes:

- Demonstrate Efficacy (Mean annual acute SBIs is significantly less than 1/subject/year)

Secondary Outcomes:

- Average serum total IgG trough concentrations prior to each infusion
- Number of infection episodes (serious and non-serious)
- Number of days out of work/school/daycare or,
- Number of days unable to perform normal daily activities due to infections
- Ability of the IMP to maintain stable, therapeutic IgG levels
- Episodes of fever

Results

A

	Age	Weight	Dose
Average	49	88kg	552mg/kg
Range	20-70	53-132kg	300-840mg/kg

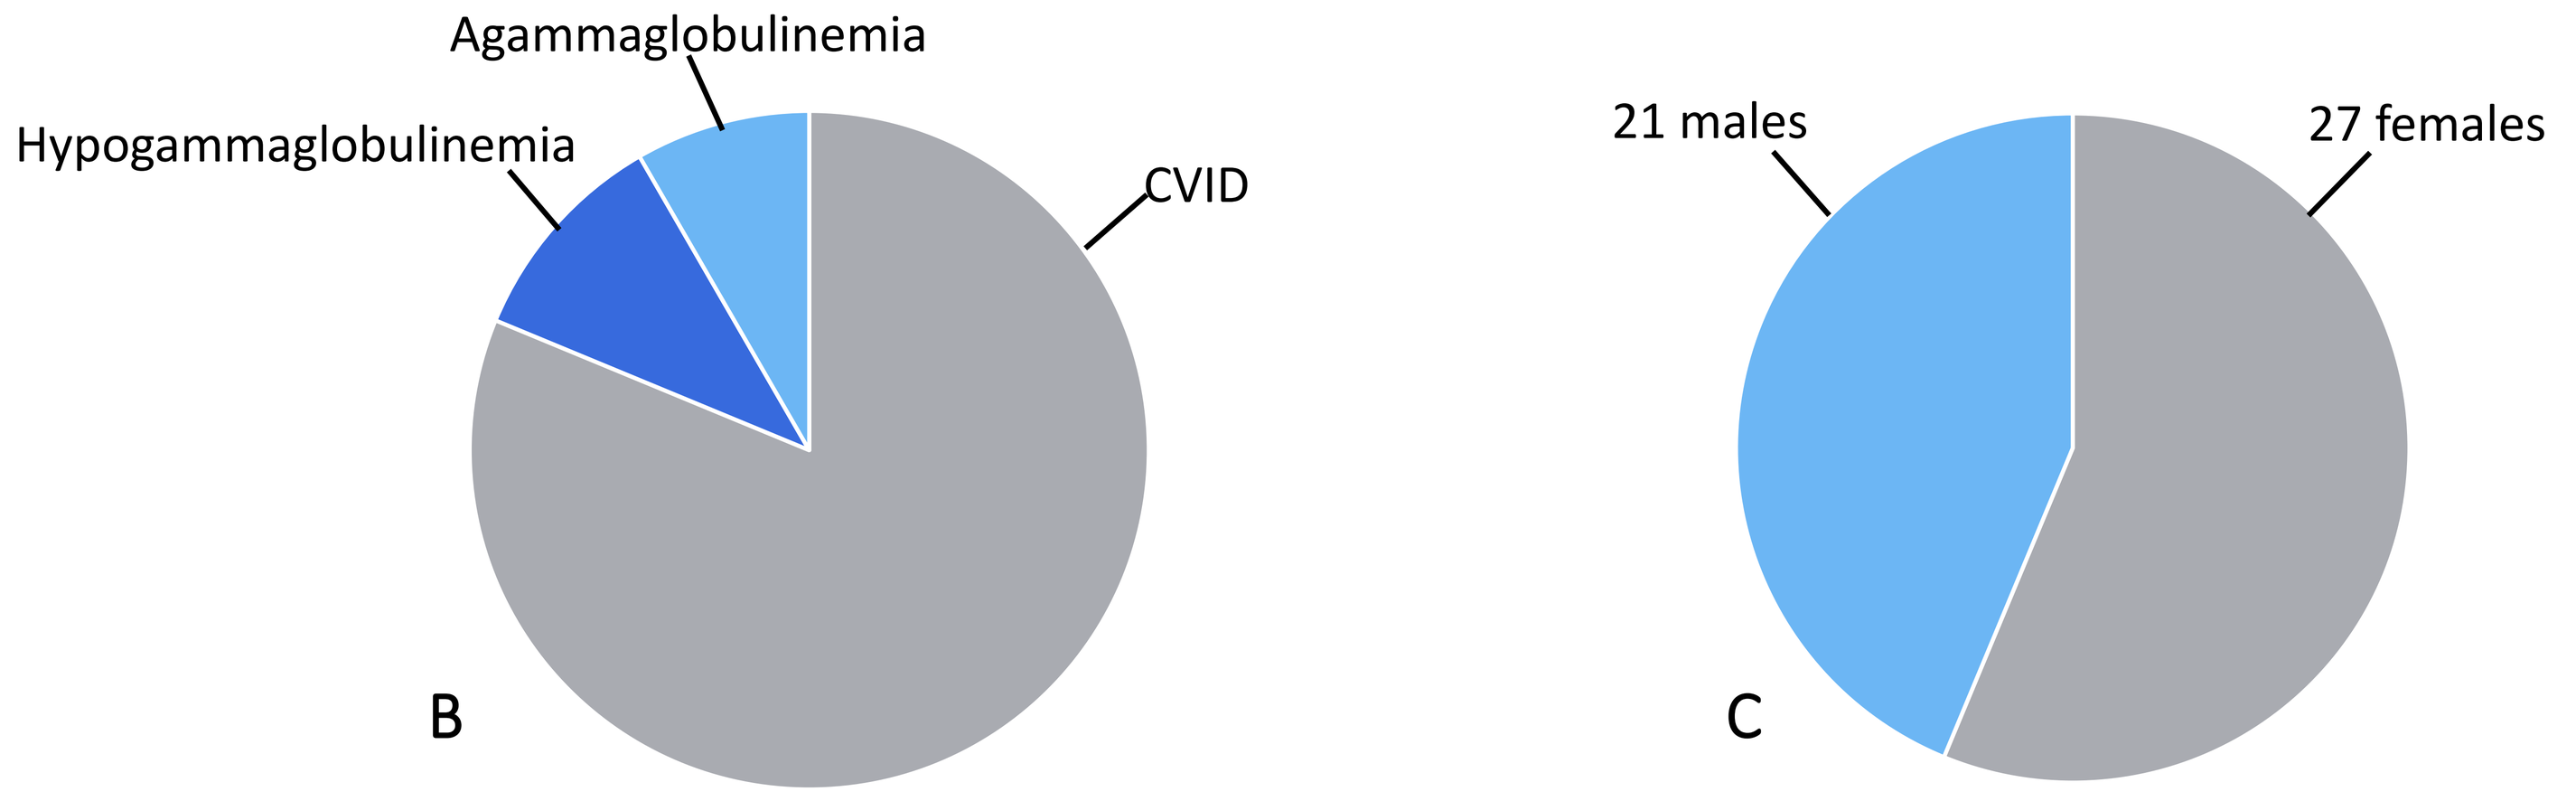
Agammaglobulinemia
Hypogammaglobulinemia
CVID
B
21 males
27 females
C

Figure 1.Enrolled Adult Patient Population data. (n= 48)Weight and Dose(A) Indication (B) and sex (C).

- At the time of the last safety review (July 2018), 48 adult subjects and 1 pediatric subject were enrolled. The adults were treated with PlasmaCap IG for a median of 162 (101-228) days; no subjects have completed the study but 5 withdrew between visits 3 and 6, due to personal reasons (n=3), generalized pruritus (n=1), and colon cancer diagnosis (n=1).

- 98.5% of the 309 infusions were completed without any administration changes, such as slowing, interrupting, or stopping the infusion.

Results (cont.)

- There were no study drug related serious adverse events (SAEs).

- The adverse reaction rate was 0.129 per infusion; almost all were mild (0.09 per infusion; n=28), and the remainder were moderate (0.035 per infusion; n=11) and severe (0.003; n=1)(**Figure 2**).

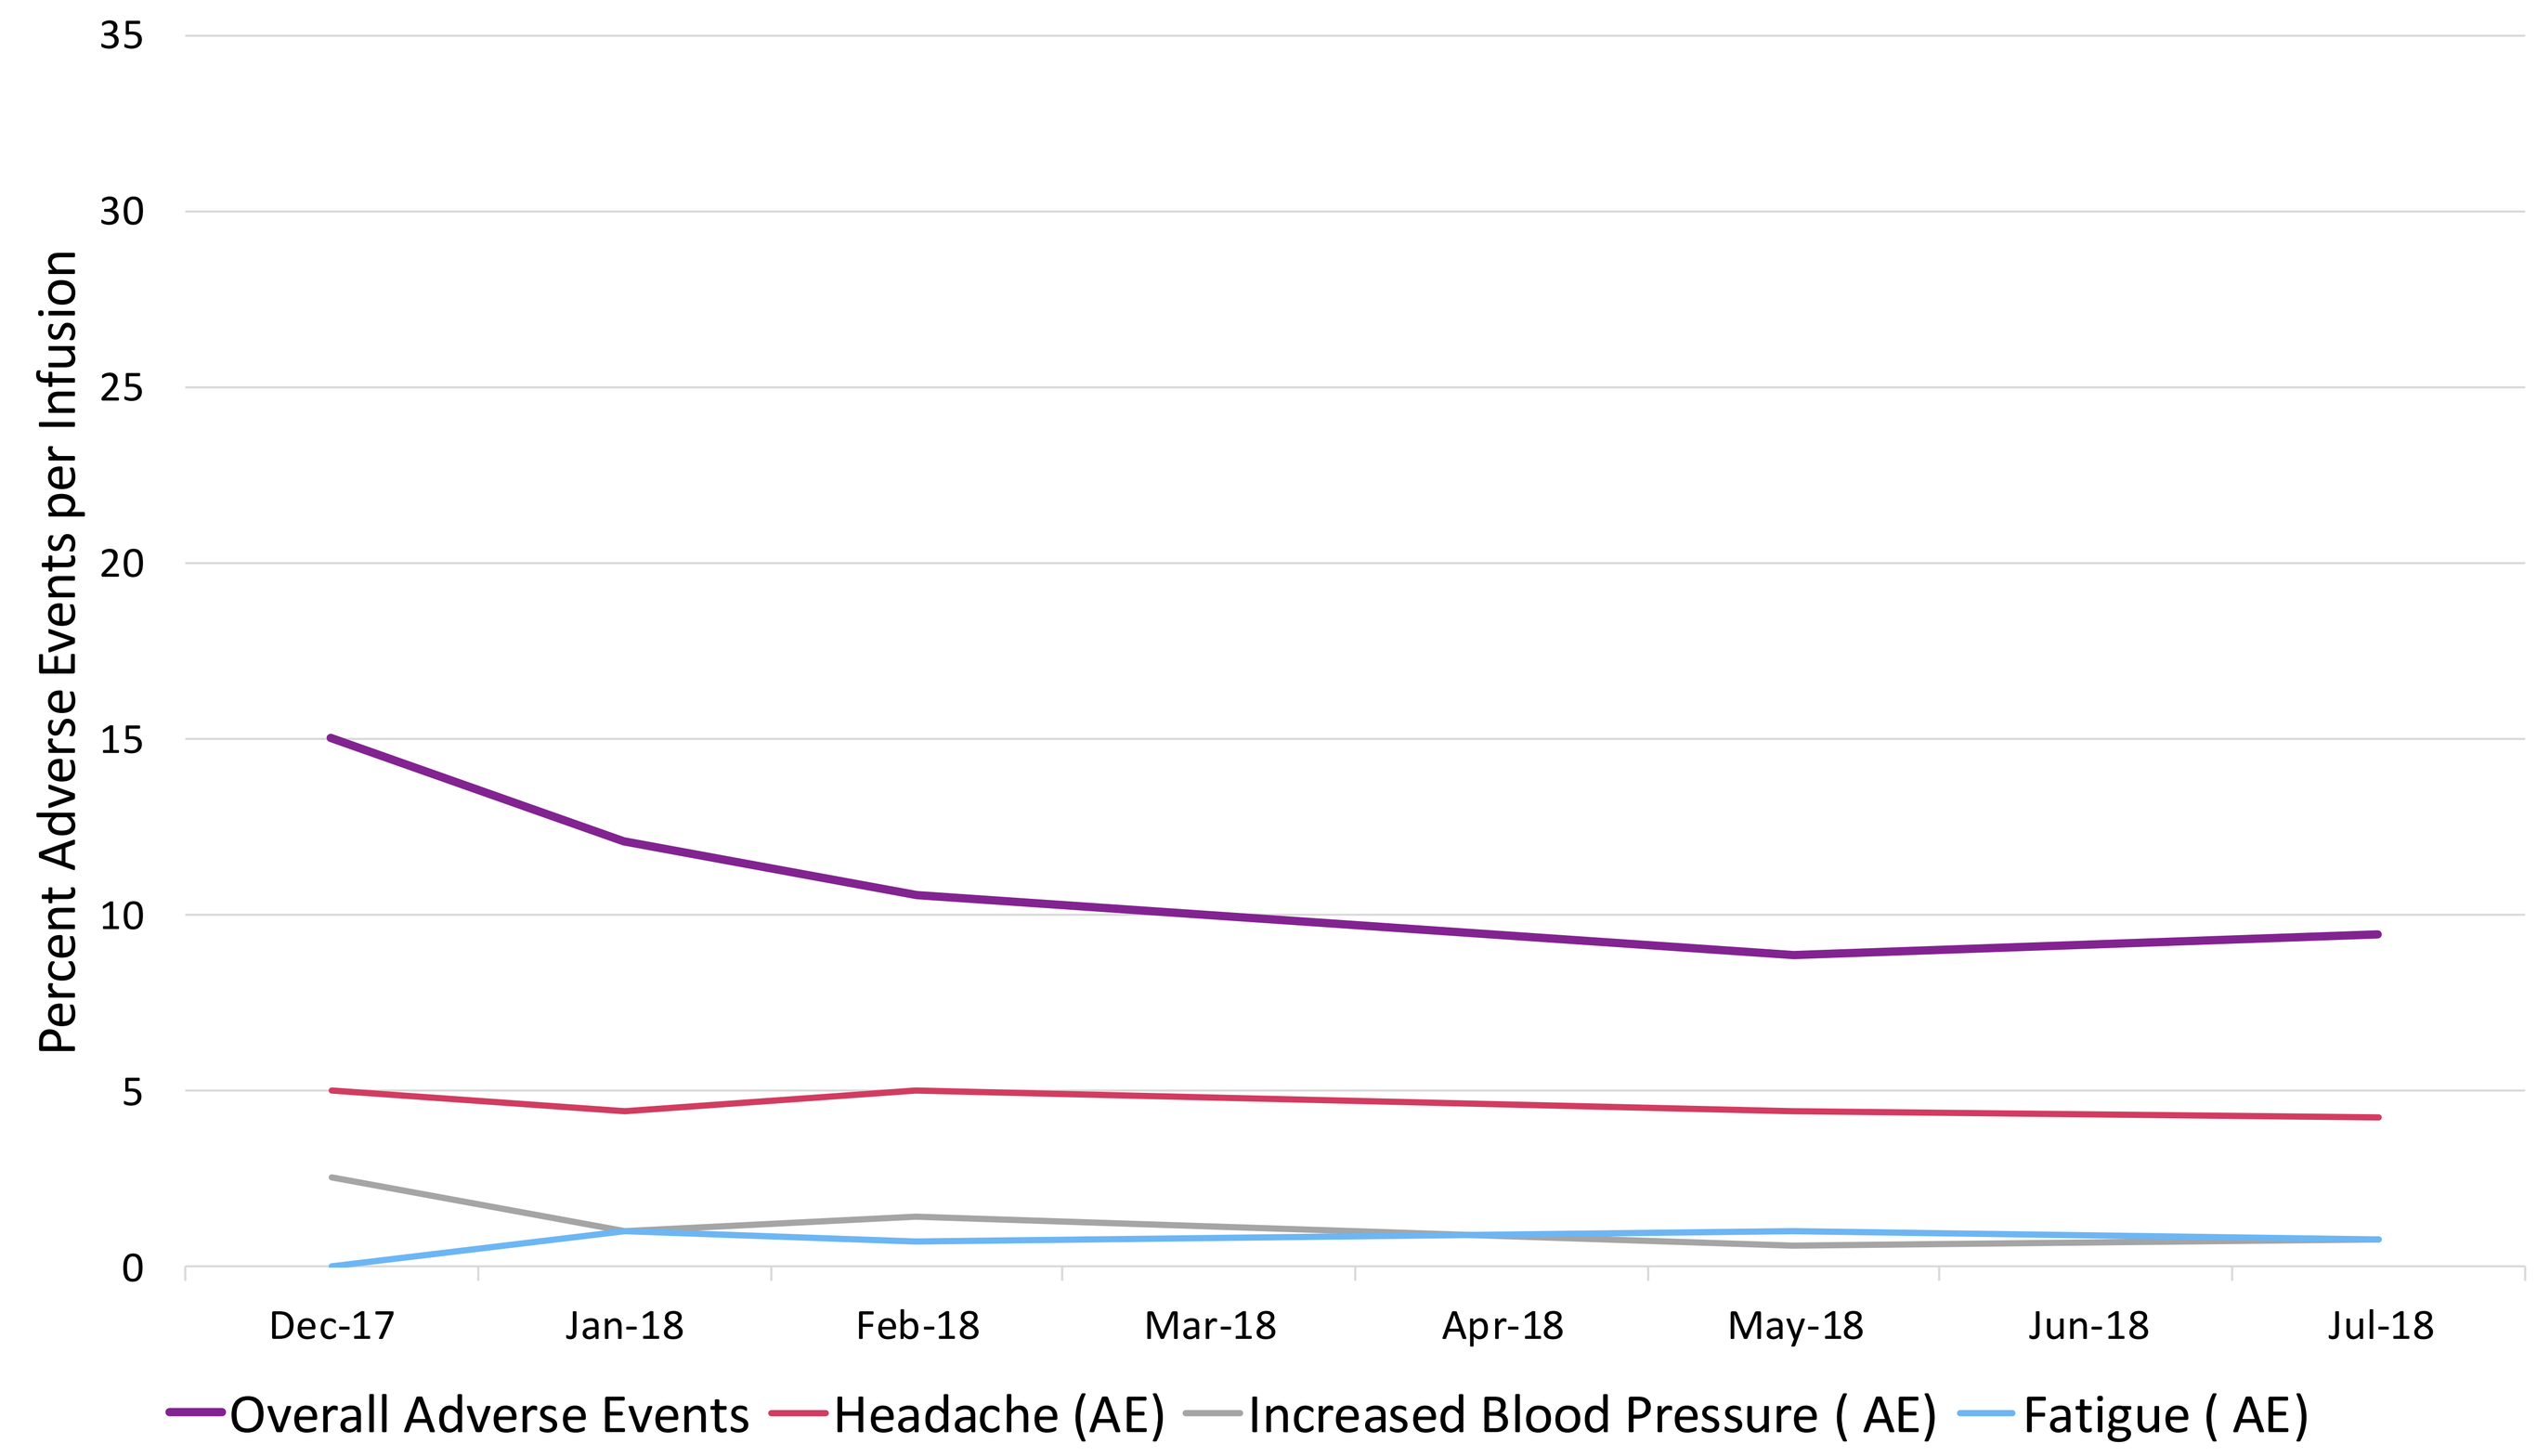
35
30
25
20
15
10
5
0
Percent Adverse Events per Infusion
Dec-17
Jan-18
Feb-18
Mar-18
Apr-18
May-18
Jun-18
Jul-18
Overall Adverse Events
Headache (AE)
Increased Blood Pressure (AE)
Fatigue (AE)

Figure 2. Percent Adverse Events in trial per PlasmaCap infusion to date. Overall AEs are in purple, among those AEs, the trend for Headache (pink), Increased Blood Pressure (grey) and Fatigue (blue)

Discussion

- The absence of any SBIs at this point in the trial suggests that PlasmaCap IG is efficacious in treating PIDD.

- Although the data is preliminary and no statistical analysis can be done, to date, the trend of Adverse Events (AE) per infusion of PlasmaCap IG appears low (**Figure 2.**). This, combined with the absence of study drug related SAEs just beyond the half way mark (63%) of the trial suggests that PlasmaCap IG is safe and well tolerated. A low rate of AEs per infusion may be a result of product purity or PlasmaCap IG's formulation's tolerability in patients. This is further highlighted with the fact that 98.5% of infusions to date were completed without any administration changes which is promising data for a 10% product.

- Further data and studies would be required to definitively conclude if PlasmaCap IG results in lower AEs while maintaining efficacy with direct comparison to other IVIG products on the market.

Conclusions

In our ongoing clinical study, these preliminary data appear to indicate PlasmaCap IG is safe and well tolerated in the treatment of patients with PIDD with no instances of related SAE to date.

References

1- Boston.com. (2018). *In the laboratory, rats are upstaging mice*. [online] Available at: http://archive.boston.com/business/articles/2010/12/27/in_the_laboratory_rats_are_upstaging_mice/ [Accessed 26 Sep. 2018].

Disclosures

All authors of this presentation are current employees of Evolve Biologics, a Division of Therapure Biopharma Inc., the commercial manufacturer using the PlasmaCap EBA™ process.