PlasmaCap EBA®: An Innovative Method of Isolating Plasma Proteins from Human Plasma

David Miller, BASc, Gillian Vanderlee BEng, MASc, Olivier Vaute, PhD, MBA, Mark Krause, BSc, MBA

Background and Rationale

- Intravenous immunoglobulin (IVIG) and subcutaneous IG (SCIG) preparations are comprised of pooled immunoglobulin G (IgG) antibodies from the plasma of thousands of donors and were initially used as an IgG replacement therapy in immunocompromised patients.[1]
- Increasingly, IG at high doses (1,000 – 2,400 mg/kg) is also used as a treatment for a variety of autoimmune and inflammatory conditions; although some autoinflammatory disorders respond to a single course of IG therapy, the majority of patients require long-term, regular infusions.[1]
- Shortages of IVIG and SCIG continue to be reported globally,[2-4] and demand for IVIG and SCIG has grown continuously since the 1980s.[5]
- The drivers for increased demand are multifactorial but are largely due to expanded indications requiring high-dose IG.[6-8]
- The manufacturing of IG is conducted by a fractionation methodology developed in the United States (US) in the 1940s known as the Cohn process or cold ethanol fractionation.[9]
- A novel protein extraction method, utilizing chromatography as the primary separation and purification process, has been developed using expanded bed adsorption (EBA) chromatography to selectively capture proteins in their native state.
- PlasmaCap IG (10% liquid formulation IVIG) is the first plasma derived product manufactured using this proprietary process.

Materials and Methods

- Evolve's PlasmaCap EBA® platform consists of a series of consecutive columns which bind a target protein, or a group of proteins, in their native state directly from cryo-poor plasma (**Figure 1**).
- Target protein(s) captured during each step of the PlasmaCap EBA® backbone can be eluted at a neutral pH in a medium ionic strength aqueous buffer (see **Figure 1**); the eluate can be concentrated, sterile filtered, and frozen for further purification, or purified immediately without an intermediate hold step.
- The five (5) key steps of the PlasmaCap EBA® process are made possible by functionalized high-density tungsten-carbide agarose beads which are suspended by upward flow (**Figure 2**)(**Table 1**).
- The upward flow results in a dynamic expanded bed which permits unclarified plasma to flow through the column with no risk of plugging, and without the need for aggressive cleaning or repacking to achieve a high cycle life.
- The PlasmaCap EBA® process was evaluated during Evolve's clinical campaign for product quality, yield, and scalability.
- In terms of scalability of the manufacturing process, it's important to note the rapid expansion of production capabilities from the laboratory-scale (LS) lots to the clinical-scale (CS) lots.

Materials and Methods (continued)

Figure 1: PlasmaCap EBA® Technology Backbone/Capture Steps and Process Flow Diagram

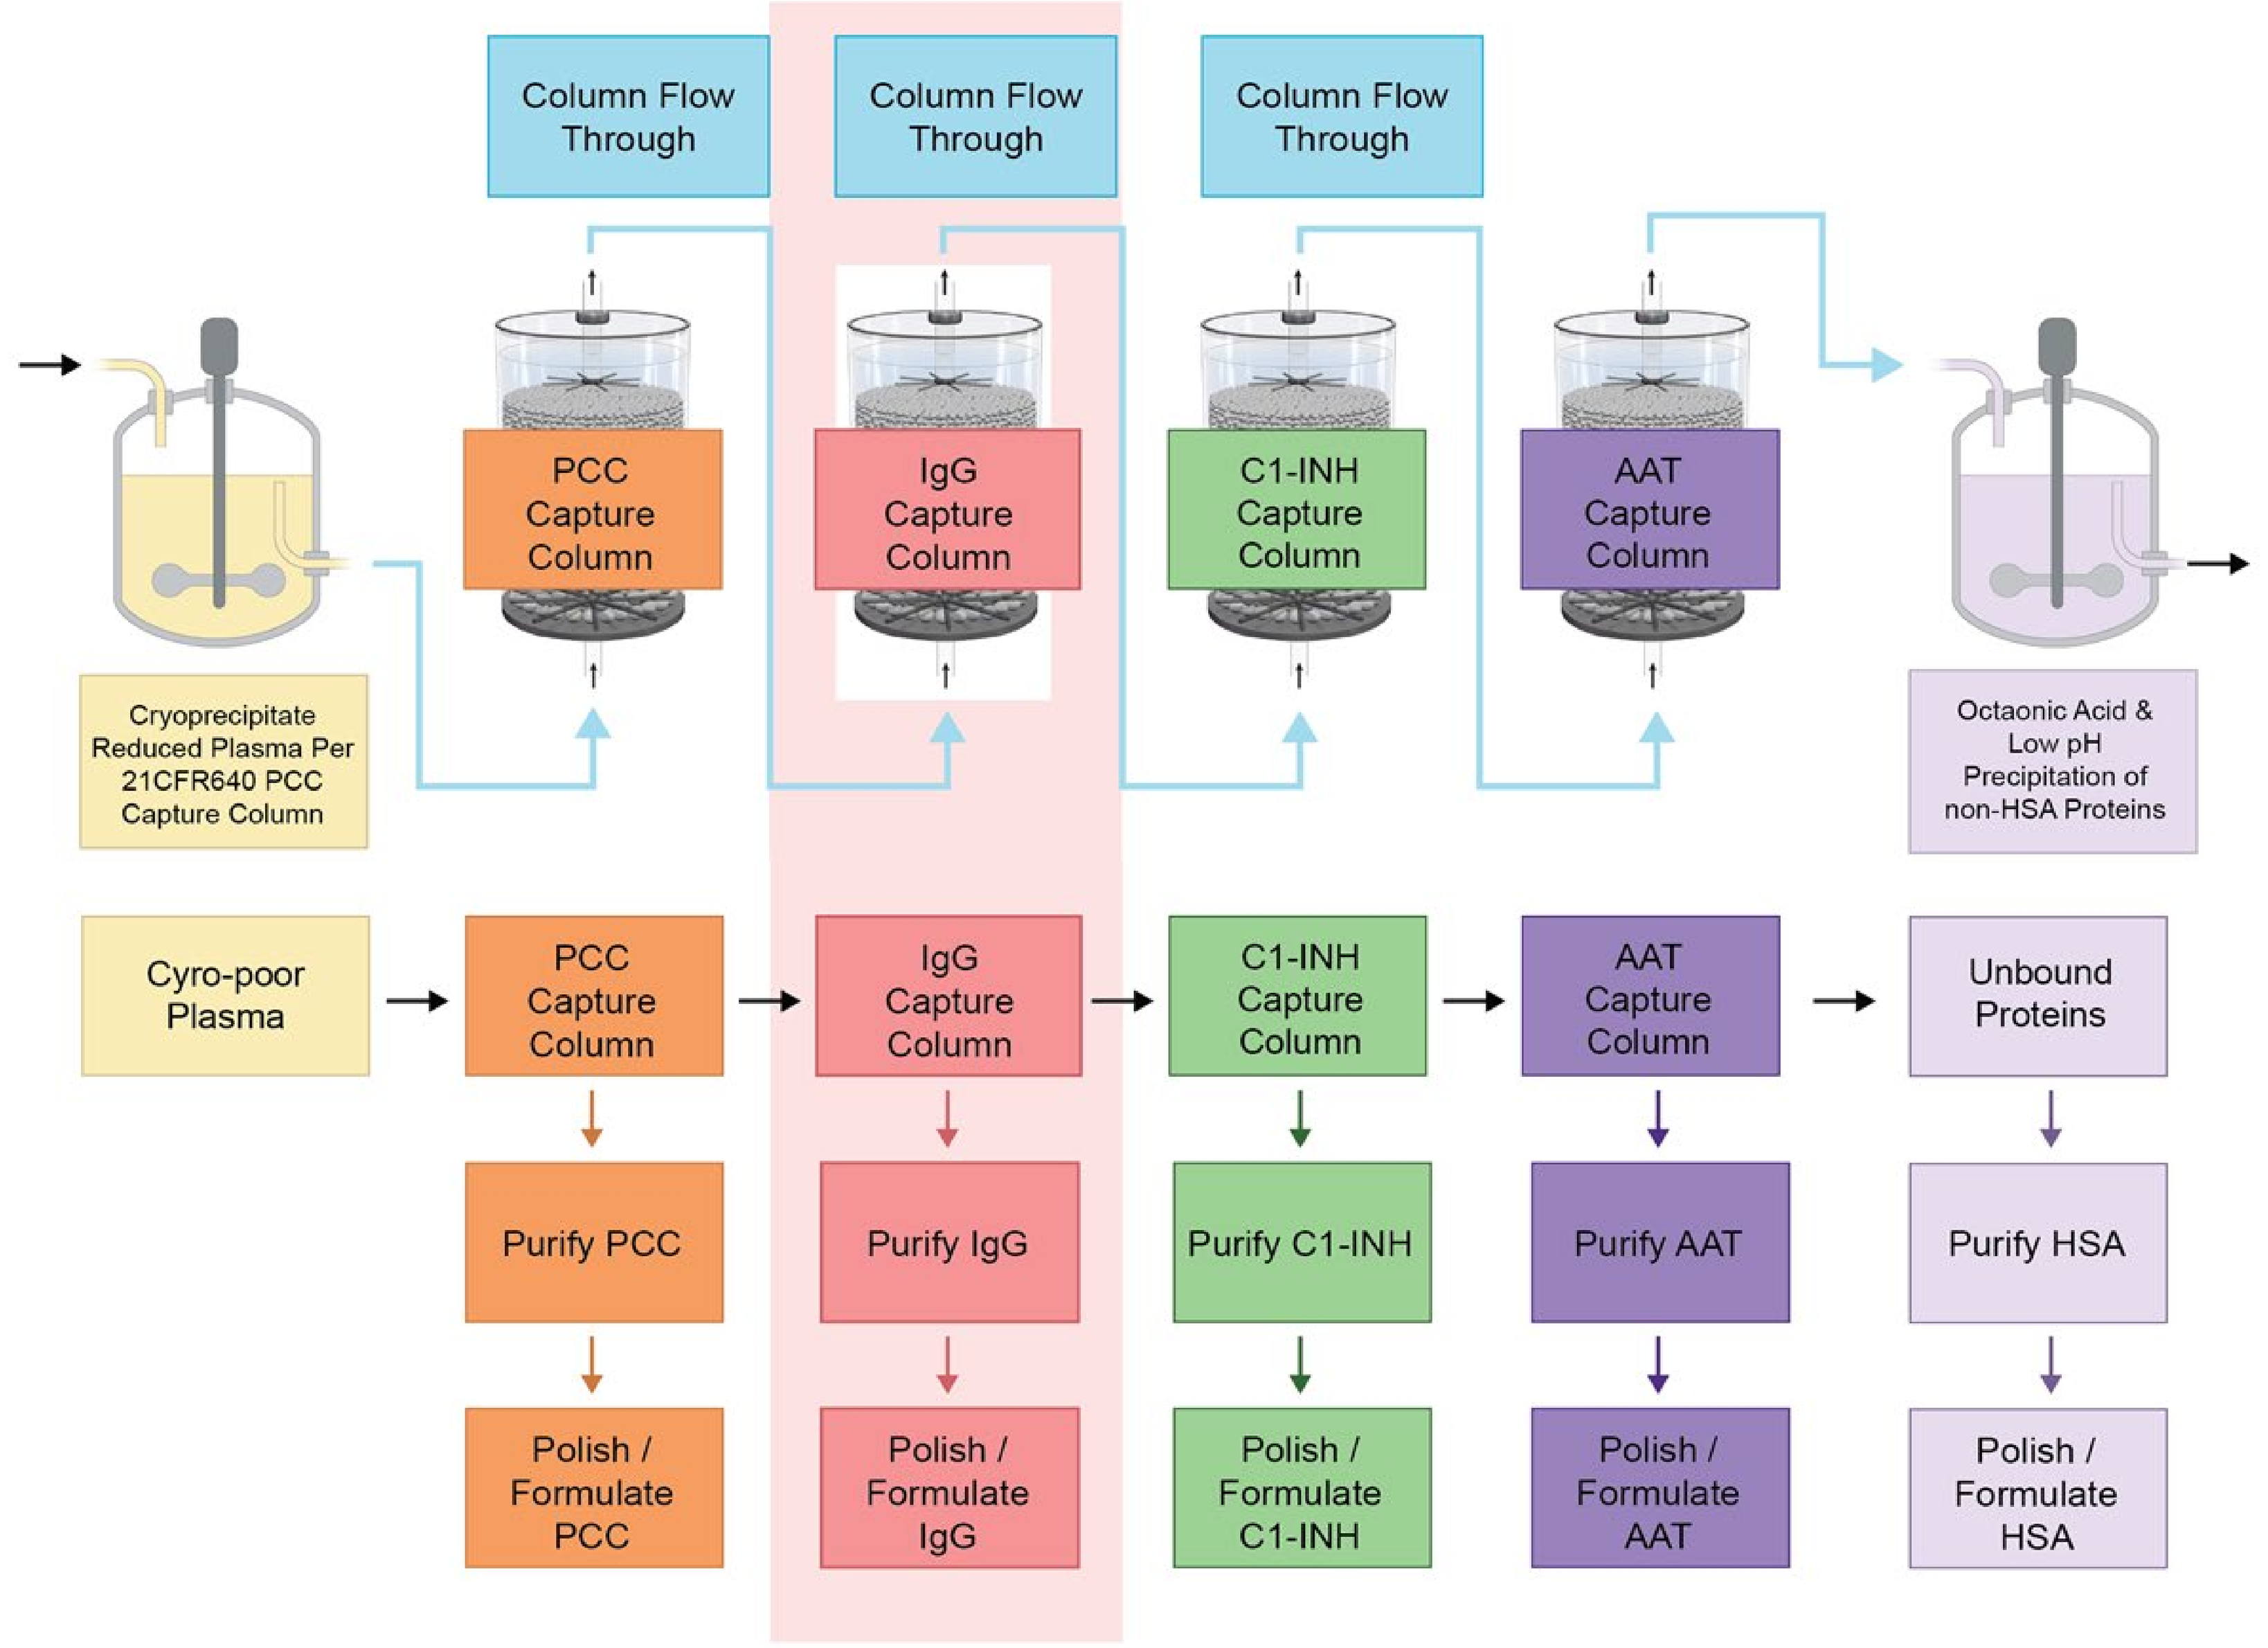

Column Flow Through
Column Flow Through
Column Flow Through
PCC Capture Column
IgG Capture Column
C1-INH Capture Column
AAT Capture Column
Cryoprecipitate Reduced Plasma Per 21CFR640 PCC Capture Column
Octanoic Acid & Low pH Precipitation of non-HSA Proteins
Cyro-poor Plasma
PCC Capture Column
IgG Capture Column
C1-INH Capture Column
AAT Capture Column
Unbound Proteins
Purify PCC
Purify IgG
Purify C1-INH
Purify AAT
Purify HSA
Polish / Formulate PCC
Polish / Formulate IgG
Polish / Formulate C1-INH
Polish / Formulate AAT
Polish / Formulate HSA

AAT=alpha-1 antitrypsin; C1-INH=C1 esterase inhibitor; HAS=human serum albumin; IgG=immunoglobulin G; PCC=prothrombin complex concentrate.

Figure 2: The Five (5) Key Steps of PlasmaCap EBA® Chromatography

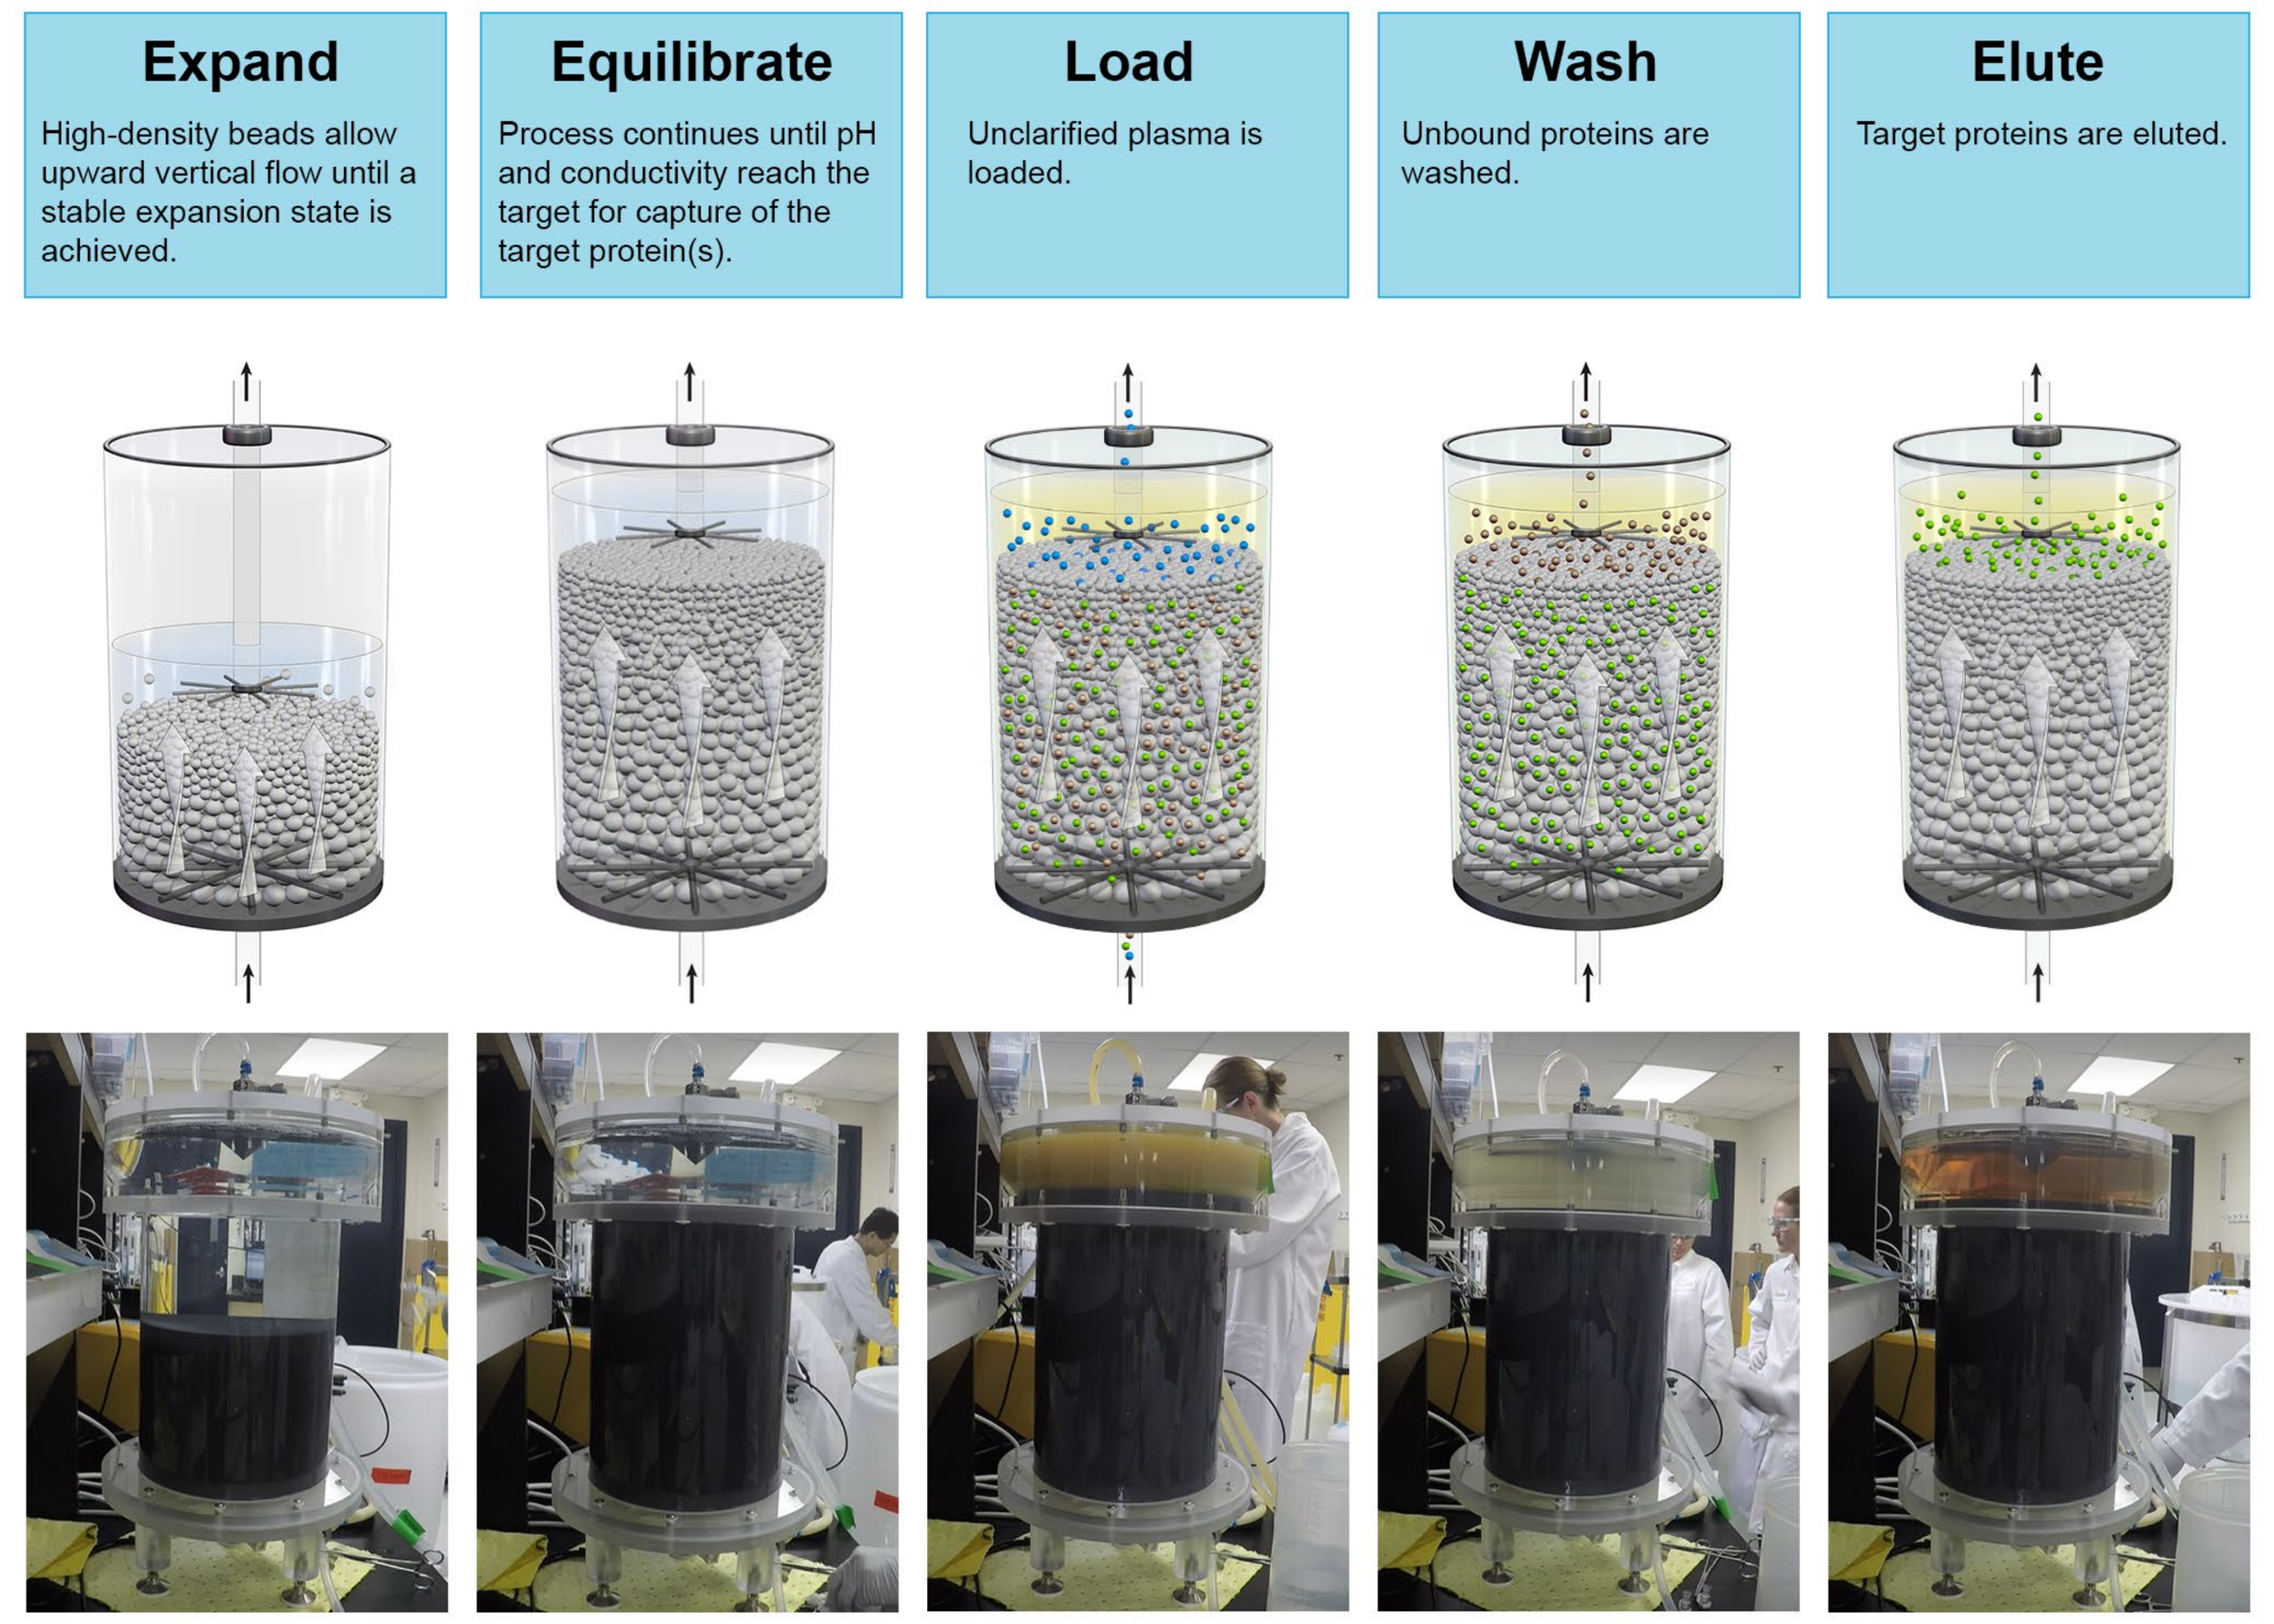

Expand
High-density beads allow upward vertical flow until a stable expansion state is achieved.
Equilibrate
Process continues until pH and conductivity reach the target for capture of the target protein(s).
Load
Unclarified plasma is loaded.
Wash
Unbound proteins are washed.
Elute
Target proteins are eluted.

Table 1: Summary of PlasmaCap EBA® Immunoglobulin Process Steps

Operation	Summary
Cryo-precipitation and recovery of cryo-poor plasma	Temperature controlled thaw followed by centrifugation to achieve selective precipitation and remove clotting factors.
Expanded bed adsorption (EBA) prothrombin complex concentrate (PCC) capture step	EBA chromatography capture step designed to bind and remove PCC from cryo-poor plasma.
Depth filtration	Depth filtration to improve performance during subsequent buffer-exchange step.
pH/conductivity adjustment	Buffer exchange by tangential flow filtration (TFF) followed by pH adjustment.
EBA immunoglobulin (IG) capture step	EBA capture chromatography to bind and isolate IG.
Low pH viral inactivation (1 of 3 viral inactivation steps) with sodium caprylate	First viral reduction step: low pH incubation in the presence of caprylate followed by depth filtration and sterile filtration.
pH/conductivity adjustment	Buffer exchange by TFF followed by sterile filtration.
Anion exchange	Polishing chromatography with packed bed anion exchange chromatography.

Results

Product Quality

- PlasmaCap EBA® technology provides several advantages over the Cohn/precipitation process.
- Key benefits are related to product quality such as lower aggregates, which are associated with increased tolerability, as well as very low levels of impurities such as Factor XIa, which may decrease the potential for thromboembolic events.
- Analysis by sodium dodecyl sulfate-polyacrylamide gel electrophoresis (SDS-PAGE) demonstrated that LS and CS lots are both highly purified.
- Specific critical quality attributes of PlasmaCap IG product produced at LS and CS are listed in **Table 2**.

Product Yield and Scalability

- In the Cohn/precipitation process, it's estimated that 40% to 50% of IG is lost in the non-IG supernatants or are coprecipitated with impurities.
- Data collected during clinical manufacturing of PlasmaCap IG (using Source plasma) has shown an average yield of 67.0% ± 5.1.
- The PlasmaCap EBA® process is expected to have the same or better yield and purity of IVIG at commercial scale production with automation and improved process flows.

Table 2: Comparison of Critical Quality Characteristics for Clinical Scale (CS) and Laboratory Scale (LS) Batches

Parameter	Limit	CS Batches (N=13	LS Batches (N=3)
Total protein	100 ± 10 g/L	100	101
Total immunoglobulin G (IgG)	100 ± 10 g/L	103	101
IgG purity	≥96%	96.9%	99.6
Molecular size	Mono- and dimeric ≥90%	99.1%	99.9
	Polymeric<2%	0.1%	<0.1
	Fragment <3%	0.7%	<0.1
IgG subclasses	IgG1 ≥55%	68%	63
	IgG2 ≥20%	28%	32
	IgG3 ≥1.5%	2.0%	3.3
	IgG4 ≥1.5%	2.2%	3.5
Immunoglobulin A (IgA)	≤100 μg/mL	17	15
Immunoglobulin M (IgM)	≤20 μg/mL	1	<0.2
Apolipoprotein H	≤1.00 mg/mL	0.4	0.1
Fibrinogen	≤500 ng/mL	0.8	0.6
Thrombin generation assay (TGA) (Factor XIa)	≤1.0 mU/mL	0.1	Not applicable (n/a)
Protein kinase A (PKA)	Not more than (NMT) 35 IU/mL 3% IgG	0.2	n/a
Anticomplement activity (ACA)	≤1 CH50 U/mg IgG	0.7	n/a
Osmolality	280 ± 15 mOs/kg	280	274
Anti-measles antibodies	Not less than (NLT) 0.6x the antibody level of NIH reference measles immune globulin	0.8	n/a
Anti-diphtheria antibodies	NLT 2U of diphtheria antitoxin/mL	10	n/a
Anti-poliomyelitis antibodies	Type 2: NLT 0.25 x reference lot Type 1: NLT 0.6 reference lot	1	n/a
Constand fragment (Fc) function	≥60% of reference material (15 and 30 mg)	102/110	n/a
Endotoxin	≤1.0 EU/mL	≤0.5	n/a

Results (continued)

Figure 3: PlasmaCap IG Characteristics/Potential Benefits

Characteristic		Differentiators / Potential Benefits
Physiological Osmolality	→	May result in higher tolerability and lower episodes of thromboembolic events.
Ultra Low Aggregates	→	Low levels of aggregates may result in higher tolerability.
Low Levels of Factor XIa	→	May result in higher tolerability and lower episodes of thromboembolic events.
Physiologic Sialyation	→	May result in potential improved efficacy through T cell independent anti-infective mechanisms and broad anti-inflammatory mechanisms.
High Levels of Monomers and Dimers	→	May result in improved functional activity and higher tolerability.
Low Levels of Immunogoblin A (IgA)	→	May result in higher tolerability for patients with known sensitivity to IgA.

Conclusions

- Shortages of immunoglobulin continue to be reported globally and demand has grown over time creating a need for improved plasma fractionation methods to provide higher yields without compromising purity and efficacy.
- A novel protein extraction method, utilizing expanded bed adsorption (EBA) chromatography, has been developed (PlasmaCap EBA®) that utilizes high-density tungsten-carbide agarose beads, suspended by upward flow, to isolate proteins.
- The PlasmaCap EBA® platform was used to successfully develop PlasmaCap IG (10% liquid formulation IVIG) for clinical investigation with proven product efficacy, safety, purity, and scalability, at a high yield.

References

1. **Perez EE, et al.** Update on the use of immunoglobulin in human disease: a review of evidence. J Allergy Clin Immunol. 2017;139:S1-46. **2. United States Food and Drug Administration (FDA)** Information About Immunoglobulin Shortage. Available at: https://www.fda.gov/vaccines-blood-biologics/safety-availability-biologics/information-about-immune-globulin-human-product-shortage. **3. Farrugia A, et al.** The growing importance of achieving national self-sufficiency in immunoglobulin in Italy. The emergence of a national imperative. Blood Transfus. 2019;17:449-458. **4. Edington HJ, et al.** Dealing with a critical national shortage – approaches to triaging immune globulin supply in pediatric hematology and oncology. Pediatr Blood Cancer. 2020;67:e28260. **5. The Marketing Research Bureau.** Introduction to the Plasma Industry [internet]. Accessed March 14, 2021. **6 Navarro RP, et al.** Considerations for the optimal use of immunoglobulin. Ann J Manag Care. 2012;18:S67-S78. **7. Kimyongur RP, et al.** The use of IVIg in the treatment of inflammatory polyneuropathies and myasthenia gravis at the Walton Centre. J R Coll Physicians Edinb. 2019;49:5-11. **8. National Blood Authority Australia NBA).** Objective 1. Secure the supply of blood and blood products. Available at: https://www.blood.gov.au/pubs/1819report/part-2-annual-performance/objective-1-secure-supply-blood-and-blood-products.html. **9. Cohn EJ, et al.** Preparation and properties of serum plasma proteins. IV. A system for the separation into fractions of the protein and lipoprotein components of biological tissues and fluids. J Am Chem Soc. 1946;62:459-75.

Acknowledgments

Evolve Biologics would like to thank Lauren Holliday for assistance with illustrations and Shireen Dunwoody of Dunwoody Consulting for assistance with poster design and development.

Presented at the: **2022 Immunoglobulin National Society (IgNS) Annual Meeting, September 29 – October 2**